ABSTRACT #1076

Anchored immunotherapy with intratumorally administered aluminum hydroxide-tethered IL-12 induces potent anti-tumor immune response

Sailaja Battula[1], Gregory Papastoitsis[1], Howard L Kaufman[1], K. Dane Wittrup[2], Michael M Schmidt[1]

[1]Ankyra Therapeutics, Boston, MA, USA; [2]Massachusetts Institute of Technology, Cambridge, MA, USA

Abstract

Interleukin-12 (IL-12) is a potent cytokine that can promote innate and adaptive anti-cancer immunity, but its clinical development has been limited by toxicity when delivered systemically [1]. Intratumoral (IT) administration can expand the therapeutic window of IL-12 and other cytokines but is in turn limited by rapid clearance from the tumor, thereby reducing efficacy, necessitating frequent administration, and increasing systemic accumulation. We recently described an approach called 'anchored immunotherapy'[2] in which an engineered IL-12 variant is complexed with the vaccine adjuvant aluminum hydroxide (alum) to form a locally retained cytokine depot that induces potent therapeutic activity in syngeneic murine tumors after only 1 or 2 IT injections. Here, we provide additional characterization of the retention kinetics and mechanism of action of alum-complexed IL-12.

Human or murine IL-12 was genetically fused to a phosphorylated alum-binding peptide (IL-12-ABP) and mixed with aluminum hydroxide to form the therapeutic complexes ANK-101 (human) or mANK-101 (mouse). Local retention and biodistribution of labeled mANK-101 after IT administration in syngeneic murine tumors was measured by SPECT imaging. Intratumoral immune changes were detected by IHC, Nanostring, and scRNA-seq to characterize mANK-101's mechanism of action. Safety of ANK-101 was assessed in cynomolgus macaques after subcutaneous injection. Statistical comparisons between groups were performed using one-way ANOVA.

ANK-101 induces IFNγ expression from human PBMCs, purified CD8+ T-cells, and NK-cells with similar potency as native IL-12. Following IT administration in murine tumor models, mANK-101 complexes are retained locally for multiple weeks while unanchored IL-12-ABP protein is cleared within hours as detected by SPECT imaging. This enhanced tumor retention corresponded to anti-tumor activity in a diverse tumor models with a range of immunogenic potential. The extended retention leads to prolonged expression of IFNγ and other pro-inflammatory cytokine and chemokines for >1 week. Gene expression and scRNA-seq analyses suggested a profound remodeling of the tumor microenvironment after mANK-101 treatment with T-cell and NK-cell activation, shifts to pro-inflammatory myeloid cell phenotypes, and increased markers of antigen presentation and co-stimulation. ANK-101 treatment resulted in an increase in CD8$^+$ T cell infiltration and phenotypic change in myeloid cells towards M1 phenotype as measured by IHC. Subcutaneous administration of ANK-101 in cynomolgus macaques was well tolerated with minimal body weight changes with the treatment. A dose dependent elevation in plasma IP-10 and minimal changes in other immune cytokines confirms the biological and mechanistic activity of ANK-101.

Anti-tumor activity of locally retained IL-12/alum is mediated by recruitment and activation of lymphocytes and myeloid immune cells. Anchored immunotherapy may represent a general approach to improve the therapeutic potential of immuno-oncology agents.

Ankyra Platform for Intratumoral Cytokine Retention

Schematic of 'Anchored Immunotherapy'

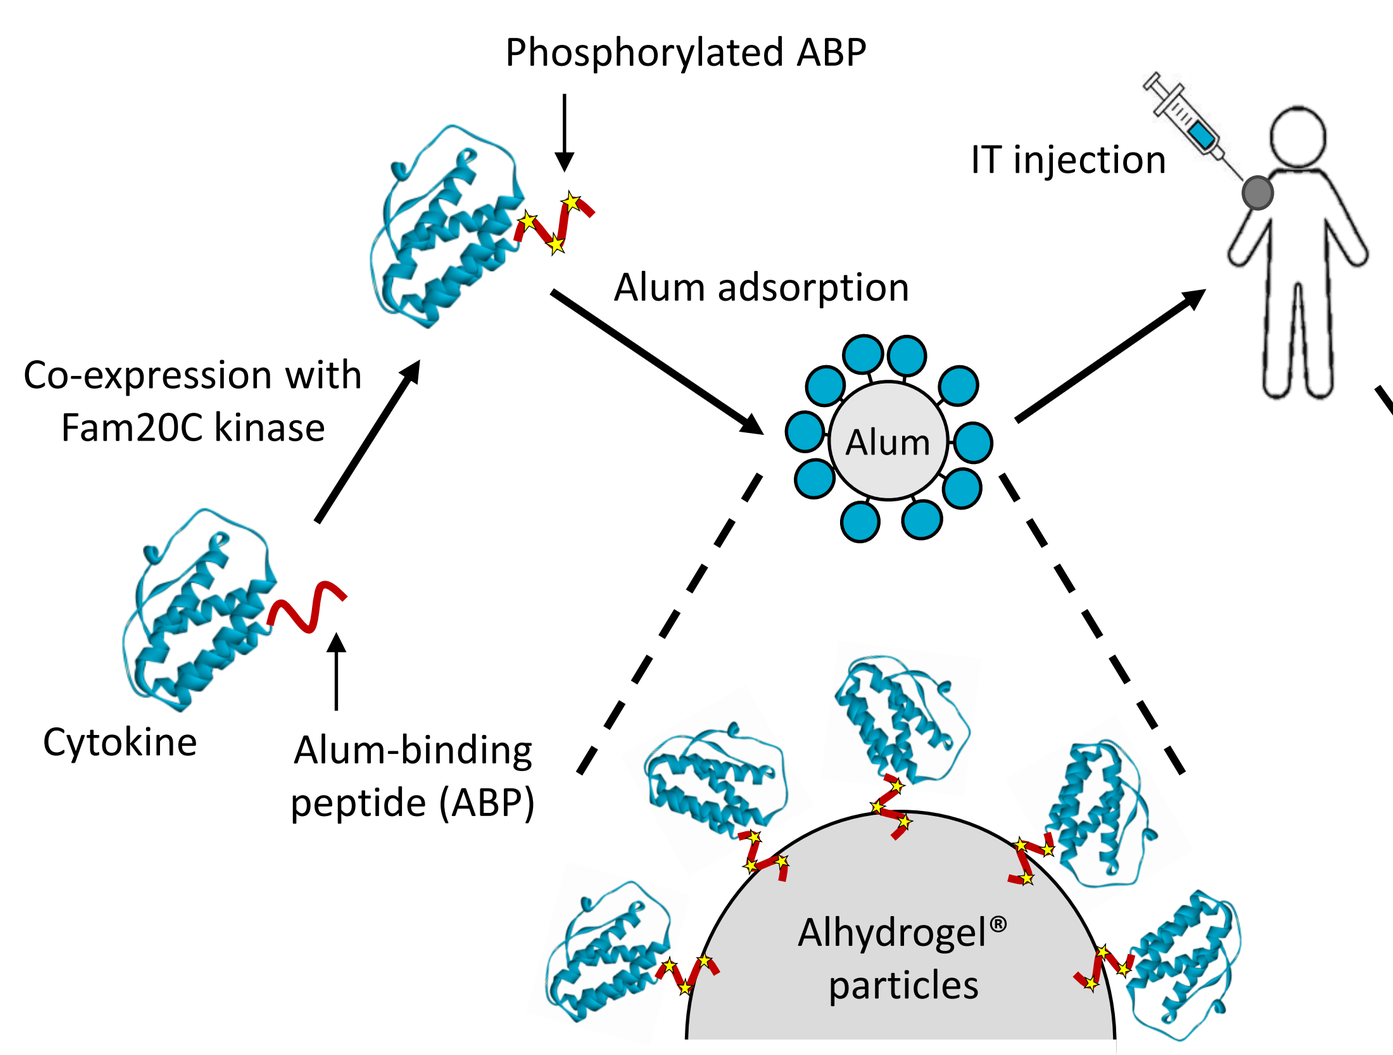

Phosphorylated ABP
IT injection
Alum adsorption
Co-expression with Fam20C kinase
Alum
Enhanced retention + efficacy
Cytokine
Alum-binding peptide (ABP)
Alhydrogel® particles

Figure 1: Cytokines and other potent immune agonists are genetically fused to a proprietary alum-binding peptide (ABP) and co-expressed with Fam20C kinase to phosphorylate the peptide at multiple serine residues. The phosphorylated fusion proteins are mixed with the FDA-approved vaccine adjuvant Alhydrogel® (aluminum hydroxide) to form a spontaneous complex through a phosphoserine mediated ligand exchange reaction. Cytokine/Alhydrogel® complexes are administered IT where they are locally retained due to their size and charge, leading to a cytokine depot that promotes potent and long-lasting local immune activation.

Figure 2: Single-chain human IL-12 with wild-type p40 and p35 sequences was genetically fused at its C-terminus to an alum-binding peptide to form IL-12-ABP, and complexed with a 10X mass excess of Alhydrogel® to form ANK-101. A mouse surrogate molecule containing the mouse IL-12 sequence (mIL-12-ABP) was also generated and complexed with Alhydrogel® and referred to as mANK-101.

ANK-101= IL-12-ABP + Alhydrogel®

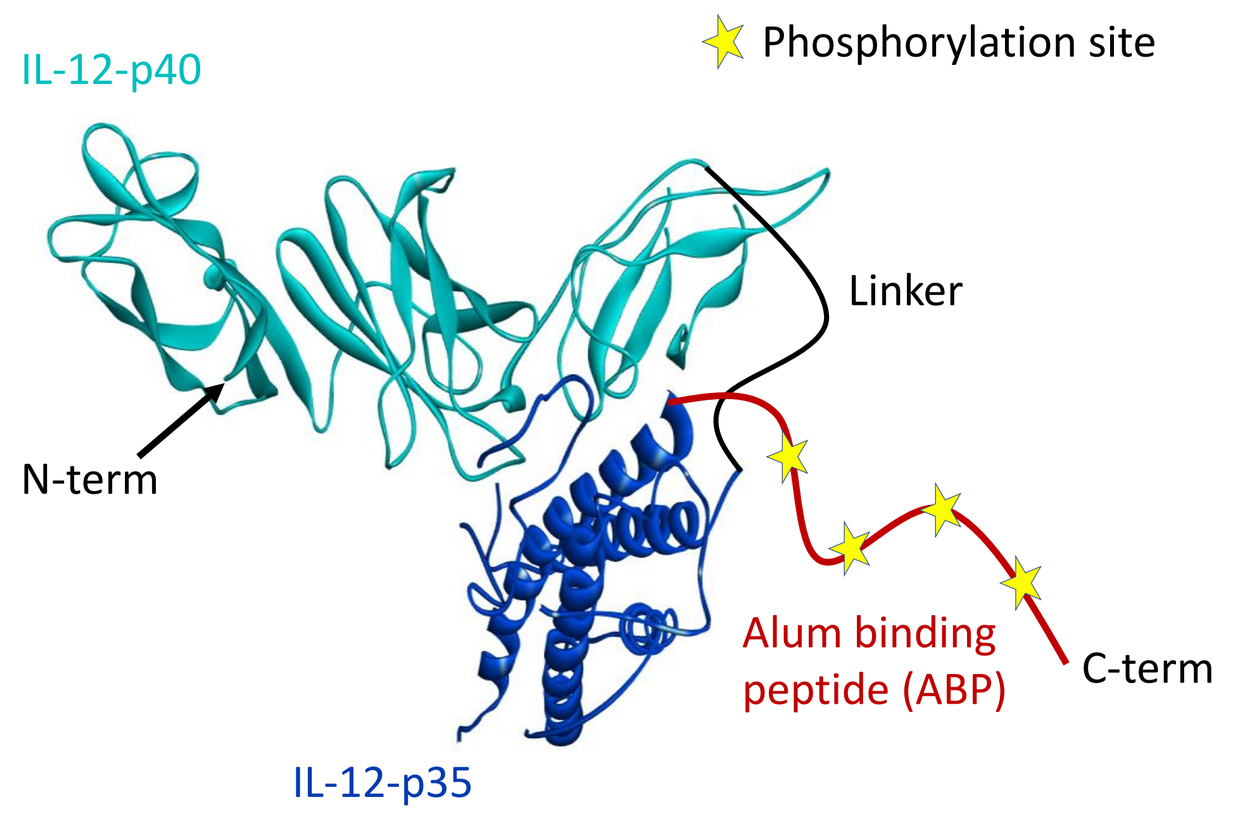

IL-12-p40
Phosphorylation site
Linker
N-term
Alum binding peptide (ABP)
C-term
IL-12-p35

ANK-101 Complex Retains IL-12 Functional Activity

Phosphorylated IL-12-ABP proteins form stable complex with Alhydrogel® particles

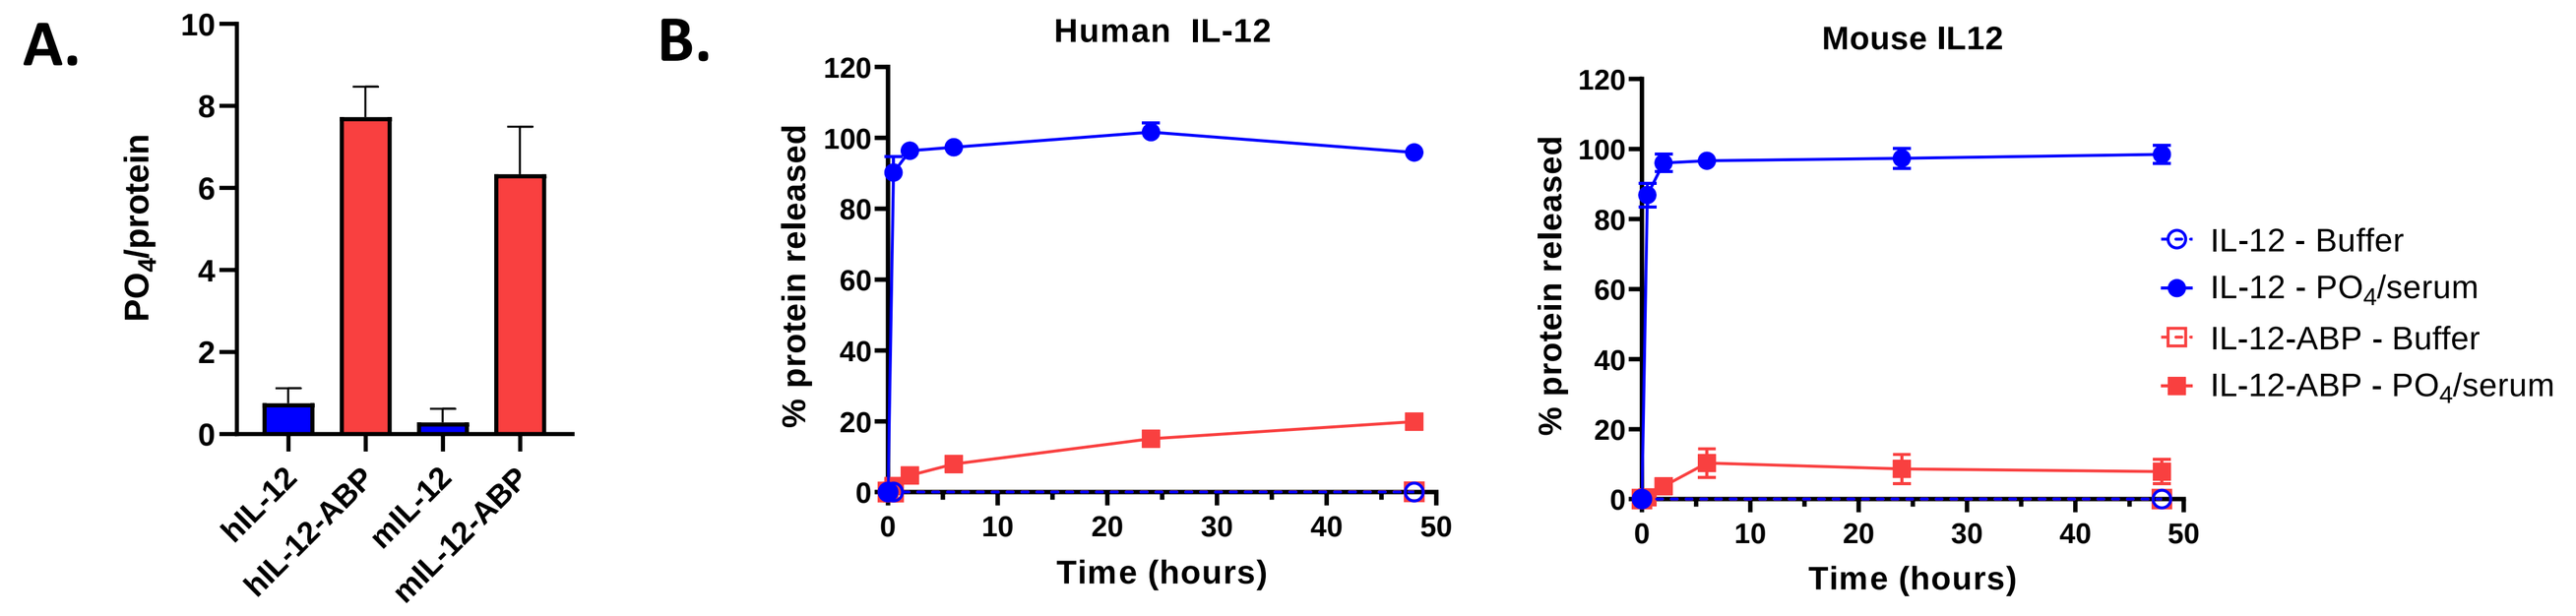

A.
PO4/protein
hIL-12
hIL-12-ABP
mIL-12
mIL-12-ABP
B.
Human IL-12
Mouse IL12
% protein released
Time (hours)
IL-12 - Buffer
IL-12 - PO4/serum
IL-12-ABP - Buffer
IL-12-ABP - PO4/serum

Figure 3: (**A**) Human and mouse IL-12-ABP are phosphorylated at multiple serines following co-expression with Fam20C kinase as measured by malachite green assay. (**B**) Human and mouse IL-12-ABP or IL-12 control proteins were complexed with Alhydrogel® then incubated in elution buffer containing 1 mM phosphate and 40% serum or a non-phosphate containing buffer control. Free IL-12 or IL-12-ABP in the supernatant was measured at various times by ELISA.

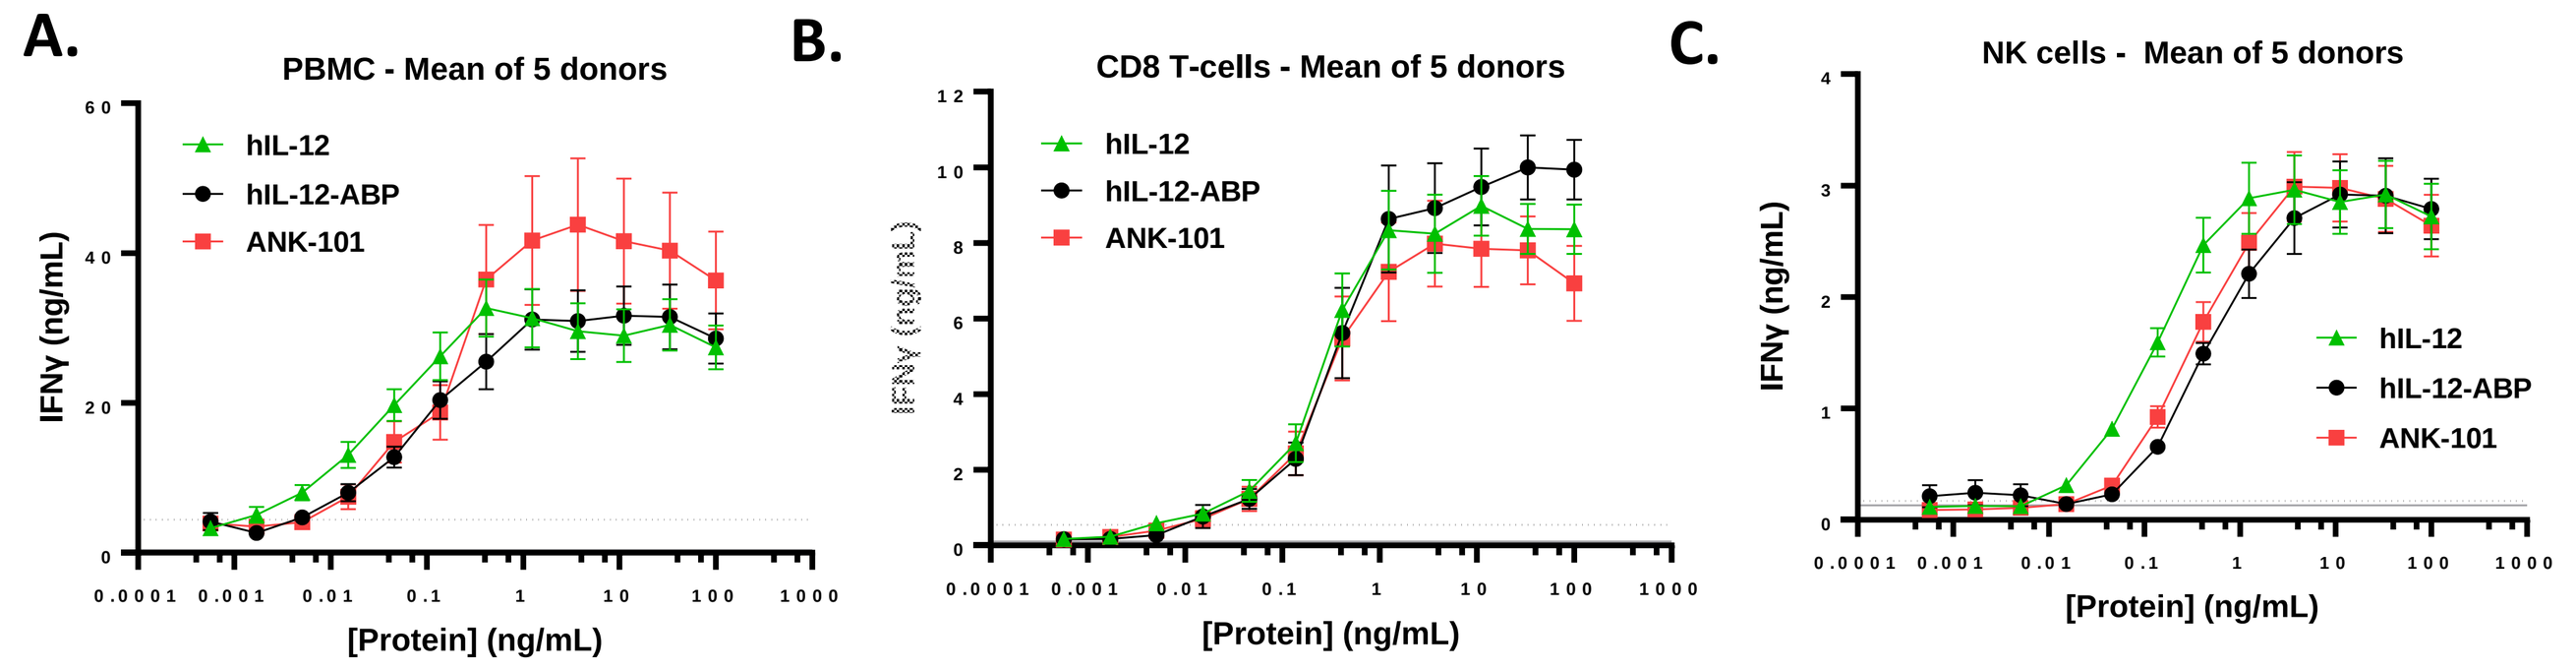

A.
PBMC - Mean of 5 donors
B.
CD8 T-cells - Mean of 5 donors
C.
NK cells - Mean of 5 donors
hIL-12
hIL-12-ABP
ANK-101
IFNγ (ng/mL)
[Protein] (ng/mL)

Figure 4: Human PBMCs (**A**) or isolated CD8$^+$ T cells (**B**) from healthy donors were stimulated with 100 ng/mL soluble anti-CD3, treated with wild-type IL-12 (hIL-12) control, free IL-12-ABP or ANK-101 complex for 3 days, and IFNγ accumulation in the culture supernatants was determined by TR-FRET. (**C**) Isolated NK cells were stimulated with soluble IL-1β (10 ng/ml) in the presence or absence of the above treatments for 3 days, and IFNγ accumulation in the culture supernatants was measured (error bars = SD).

mANK-101 is Locally Retained After IT Administration

Increased tumor retention and decreased systemic exposure of mANK-101 compared to free IL-12-ABP

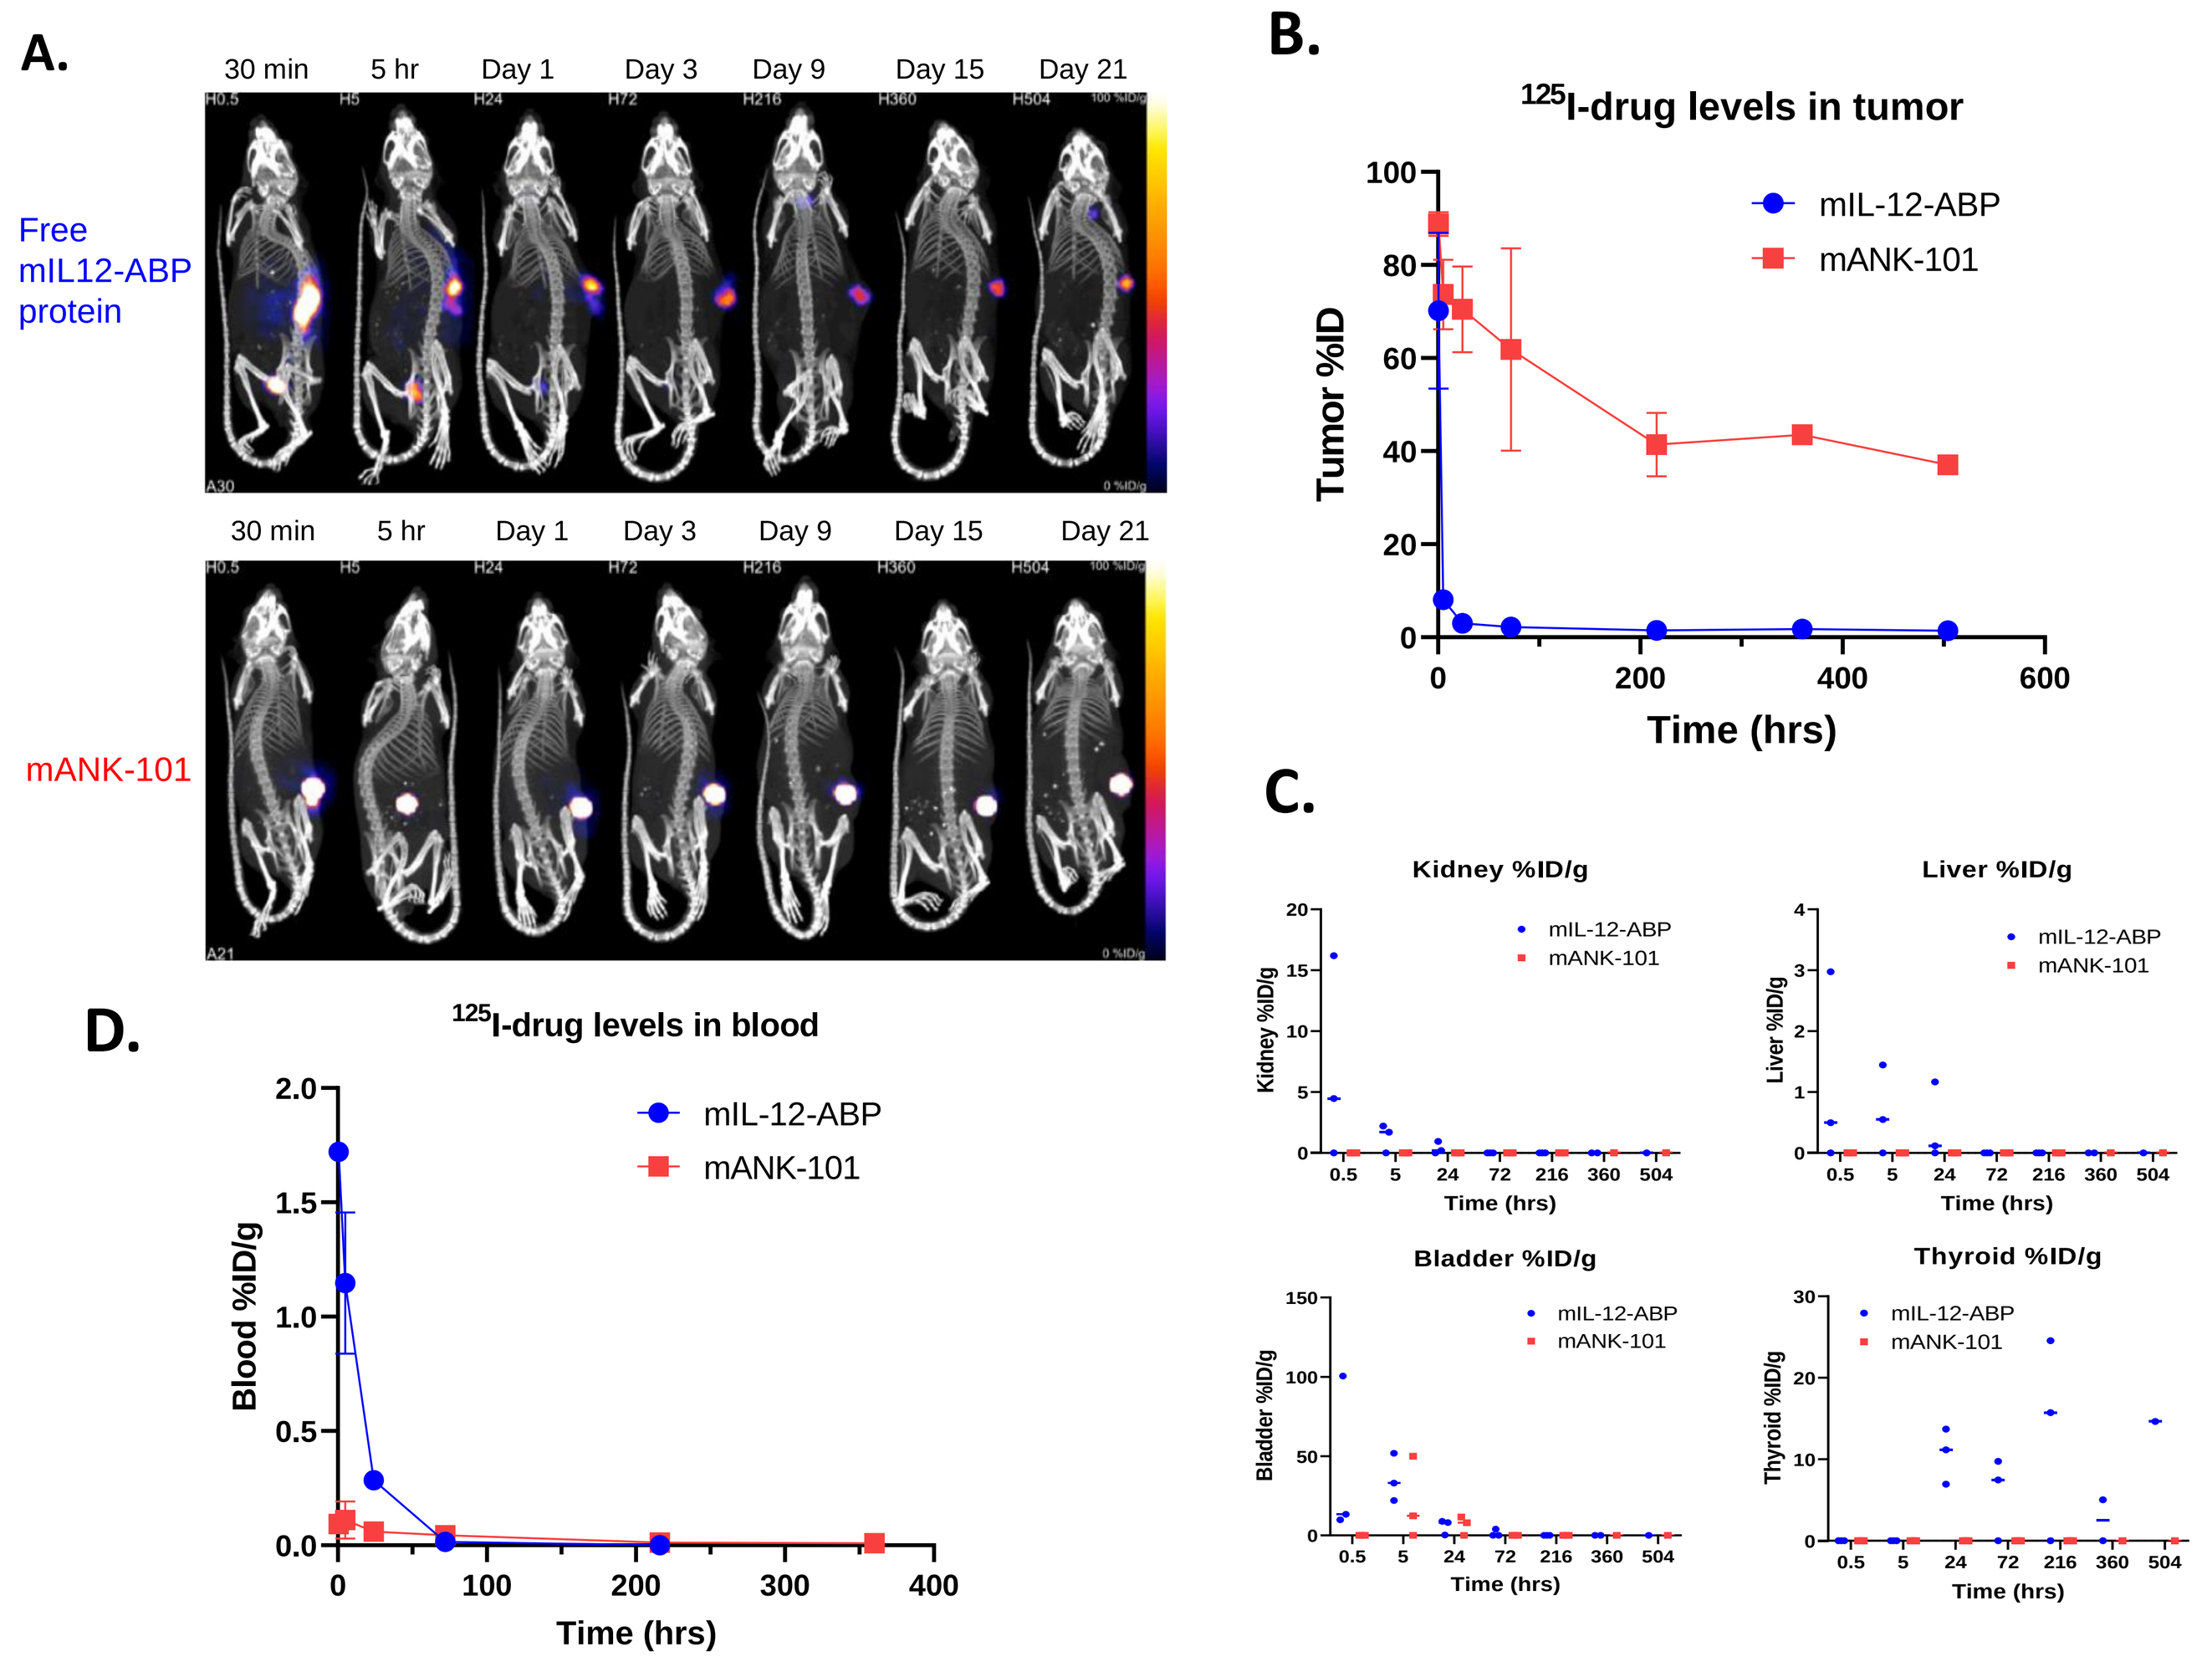

A.
30 min
5 hr
Day 1
Day 3
Day 9
Day 15
Day 21
Free mIL12-ABP protein
mANK-101
B.
125I-drug levels in tumor
Tumor %ID
Time (hrs)
mIL-12-ABP
mANK-101
C.
Kidney %ID/g
Liver %ID/g
Bladder %ID/g
Thyroid %ID/g
D.
125I-drug levels in blood
Blood %ID/g

Figure 5: 5 µg of ^{125}I labeled mIL-12-ABP was injected intratumorally in BALB/c mice bearing CT26 tumors (n = 3) either as free protein or complexed with Alhydrogel® to form mANK-101 complex, and the animals were imaged by Single-Photon Emission Computed Tomography (SPECT) over time. **(A)** Representative SPECT time course images, consistent across all 3 animals per group. The concentration of ^{125}I-mIL-12-ABP in the tumor **(B)** or peripheral tissues **(C)** was quantified over time from the SPECT images and reported as % injected dose (%ID). (**D**) Whole blood was collected at various times and radiation quantified *ex vivo* by gamma count.

mANK-101 is Efficacious in Diverse Tumor Models

Efficacy of ANK-101 in syngeneic models with varying immunogenic potential and tumor sizes

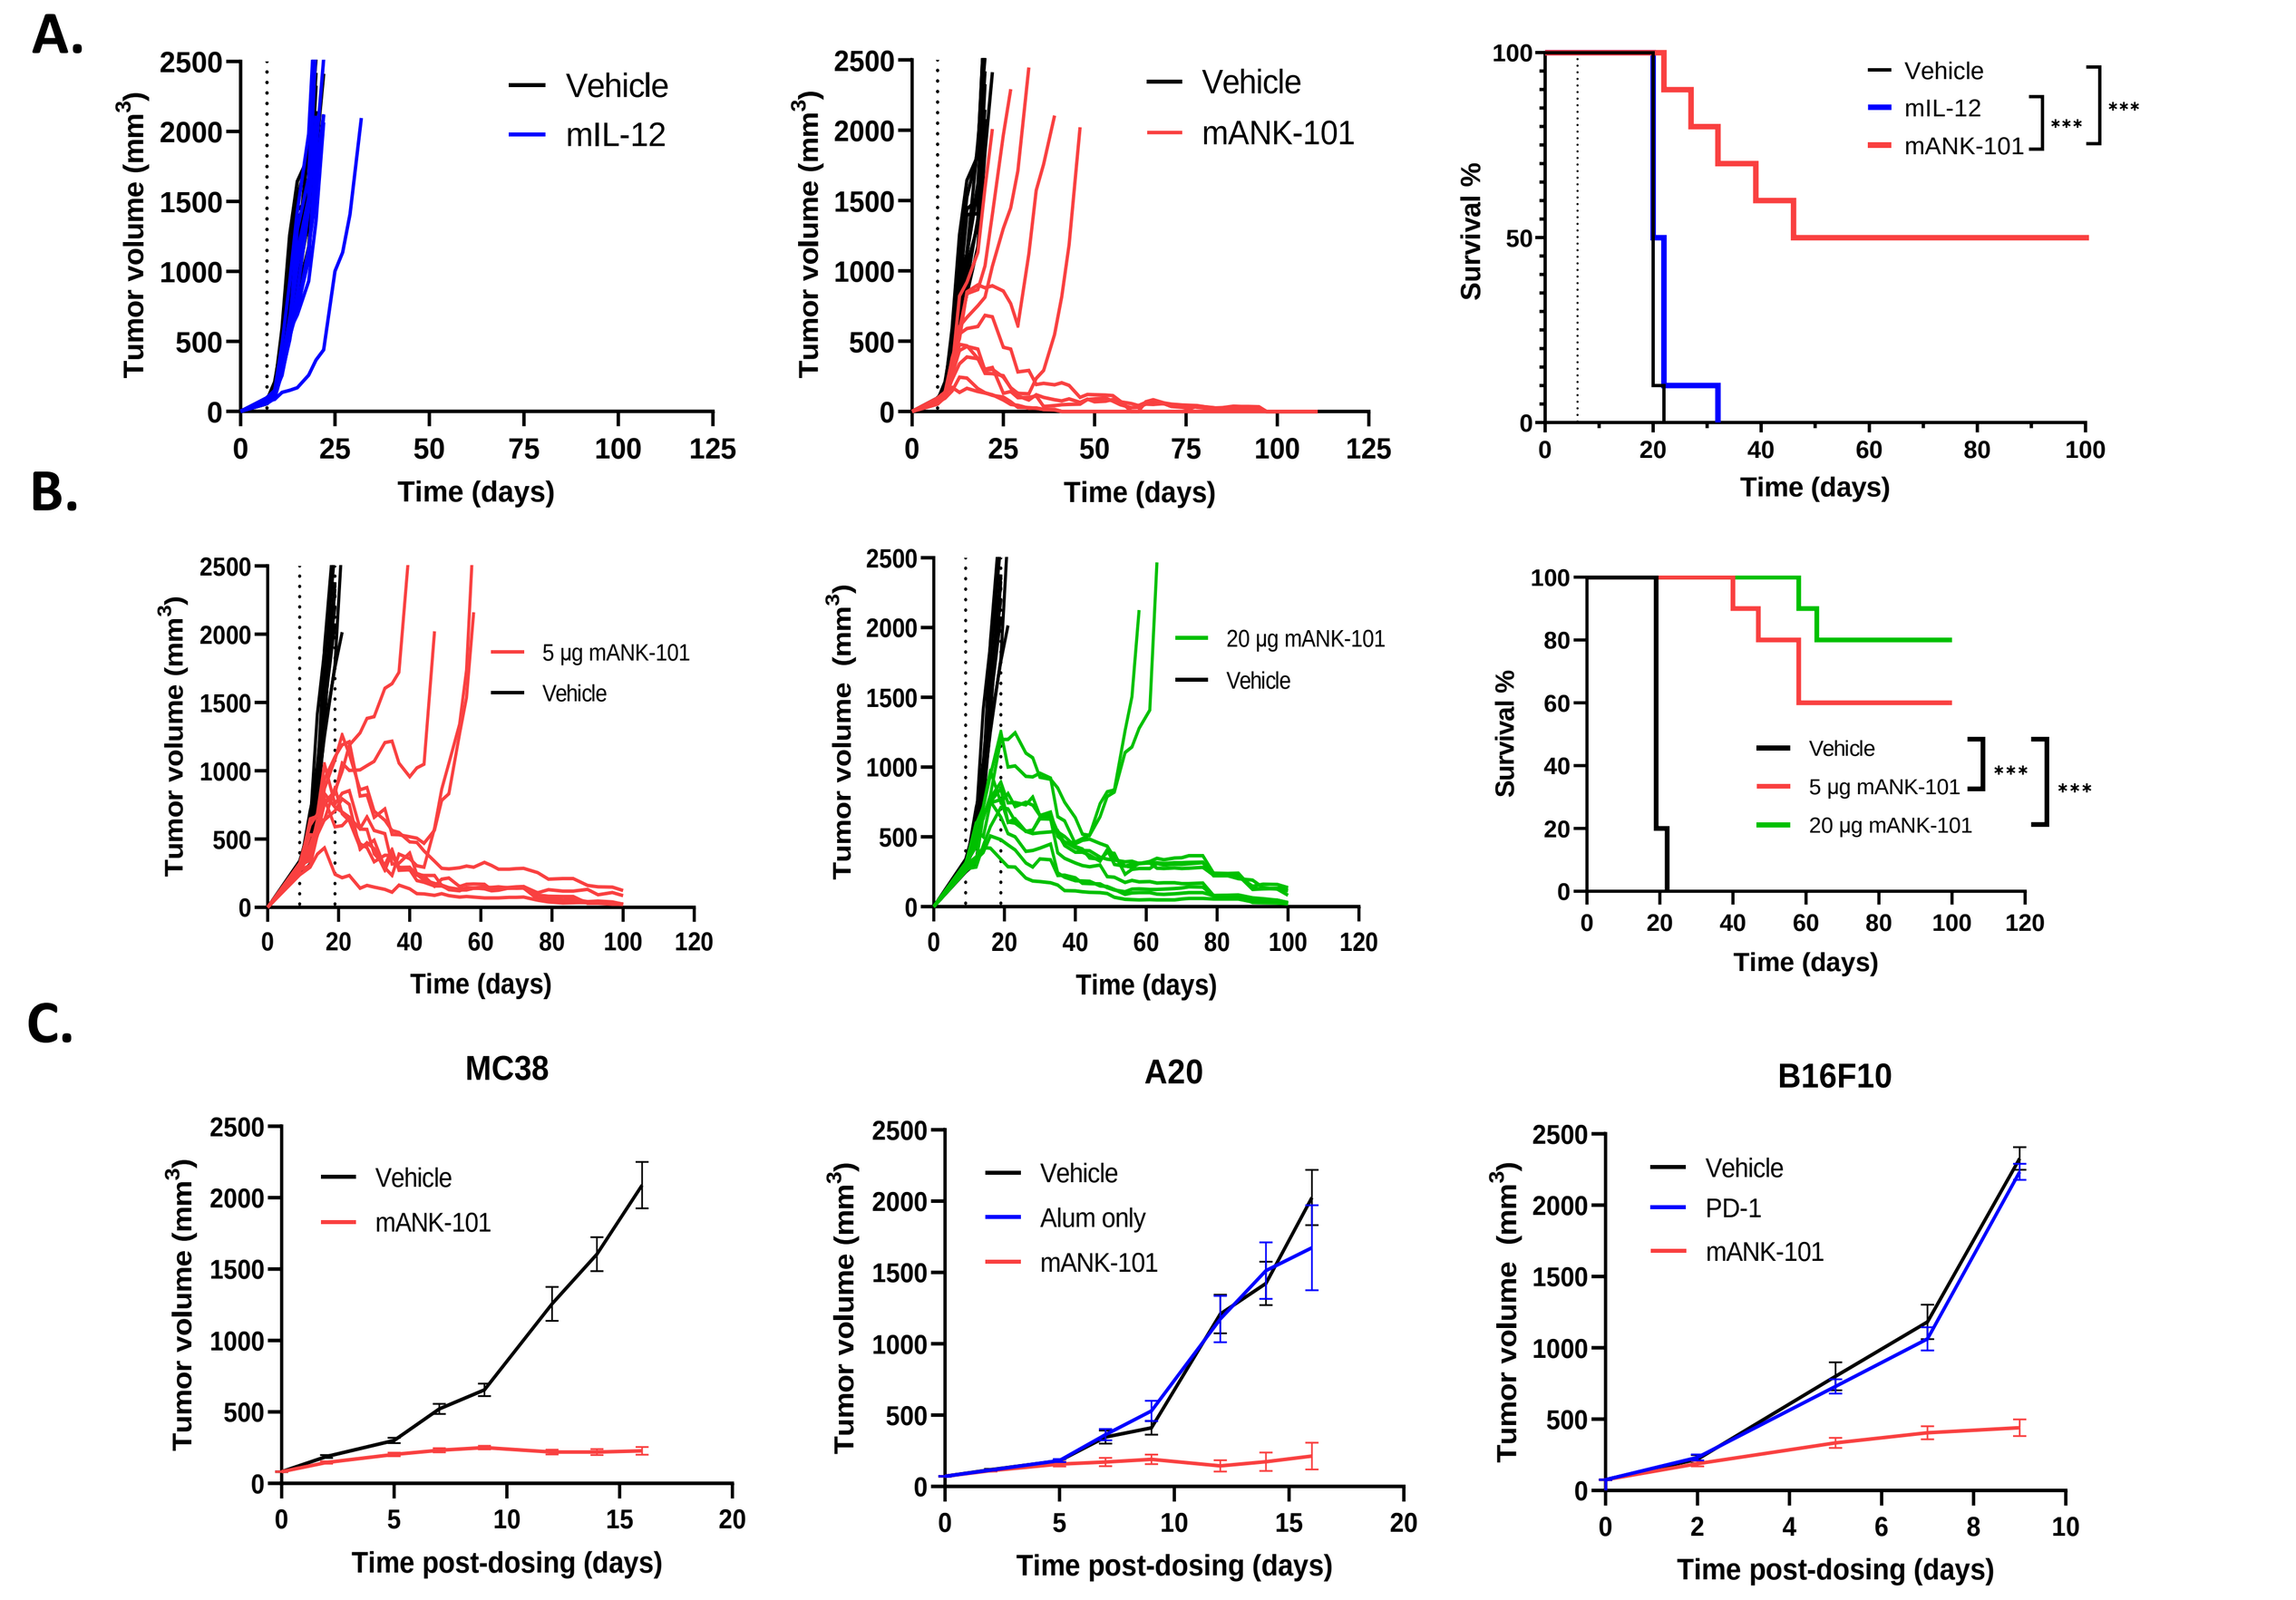

A.
Vehicle
mIL-12
mANK-101
Tumor volume (mm3)
Time (days)
Survival %
B.
5 µg mANK-101
20 µg mANK-101
C.
MC38
A20
B16F10
Alum only
PD-1
Time post-dosing (days)

Figure 6: (**A**) Tumor volume (TV) and survival for BALB/c mice (10/group) bearing ~80 mm^3 CT26 tumors that were administered a single IT injection of vehicle, 5 µg mIL-12, or 5 µg mANK-101. Mice were euthanized when TV > 2000 mm^3 (**B**) Tumor growth curves and survival of BALB/c mice bearing ~300 mm^3 CT26 tumors administered two IT doses of vehicle, 5 µg or 20 µg mANK-101 ten days apart. Mice were euthanized when TV > 2000 mm^3. (**C**) 5 µg mANK-101 was tested for anti-tumor response in various syngeneic tumor models. mANK-101 was administered as a single IT dose in the MC38 model or two IT doses 7 days apart in the A20 and B16F10 models. Alum alone has no effect on reducing tumor burden. ANK-101 is effective in PD-1 resistant B16F10 model. (***P < 0.0001, log rank test)

mANK-101 Modulates both Innate and Adaptive Immunity

Immune modulatory effects confirmed both by gene and protein expression analyses

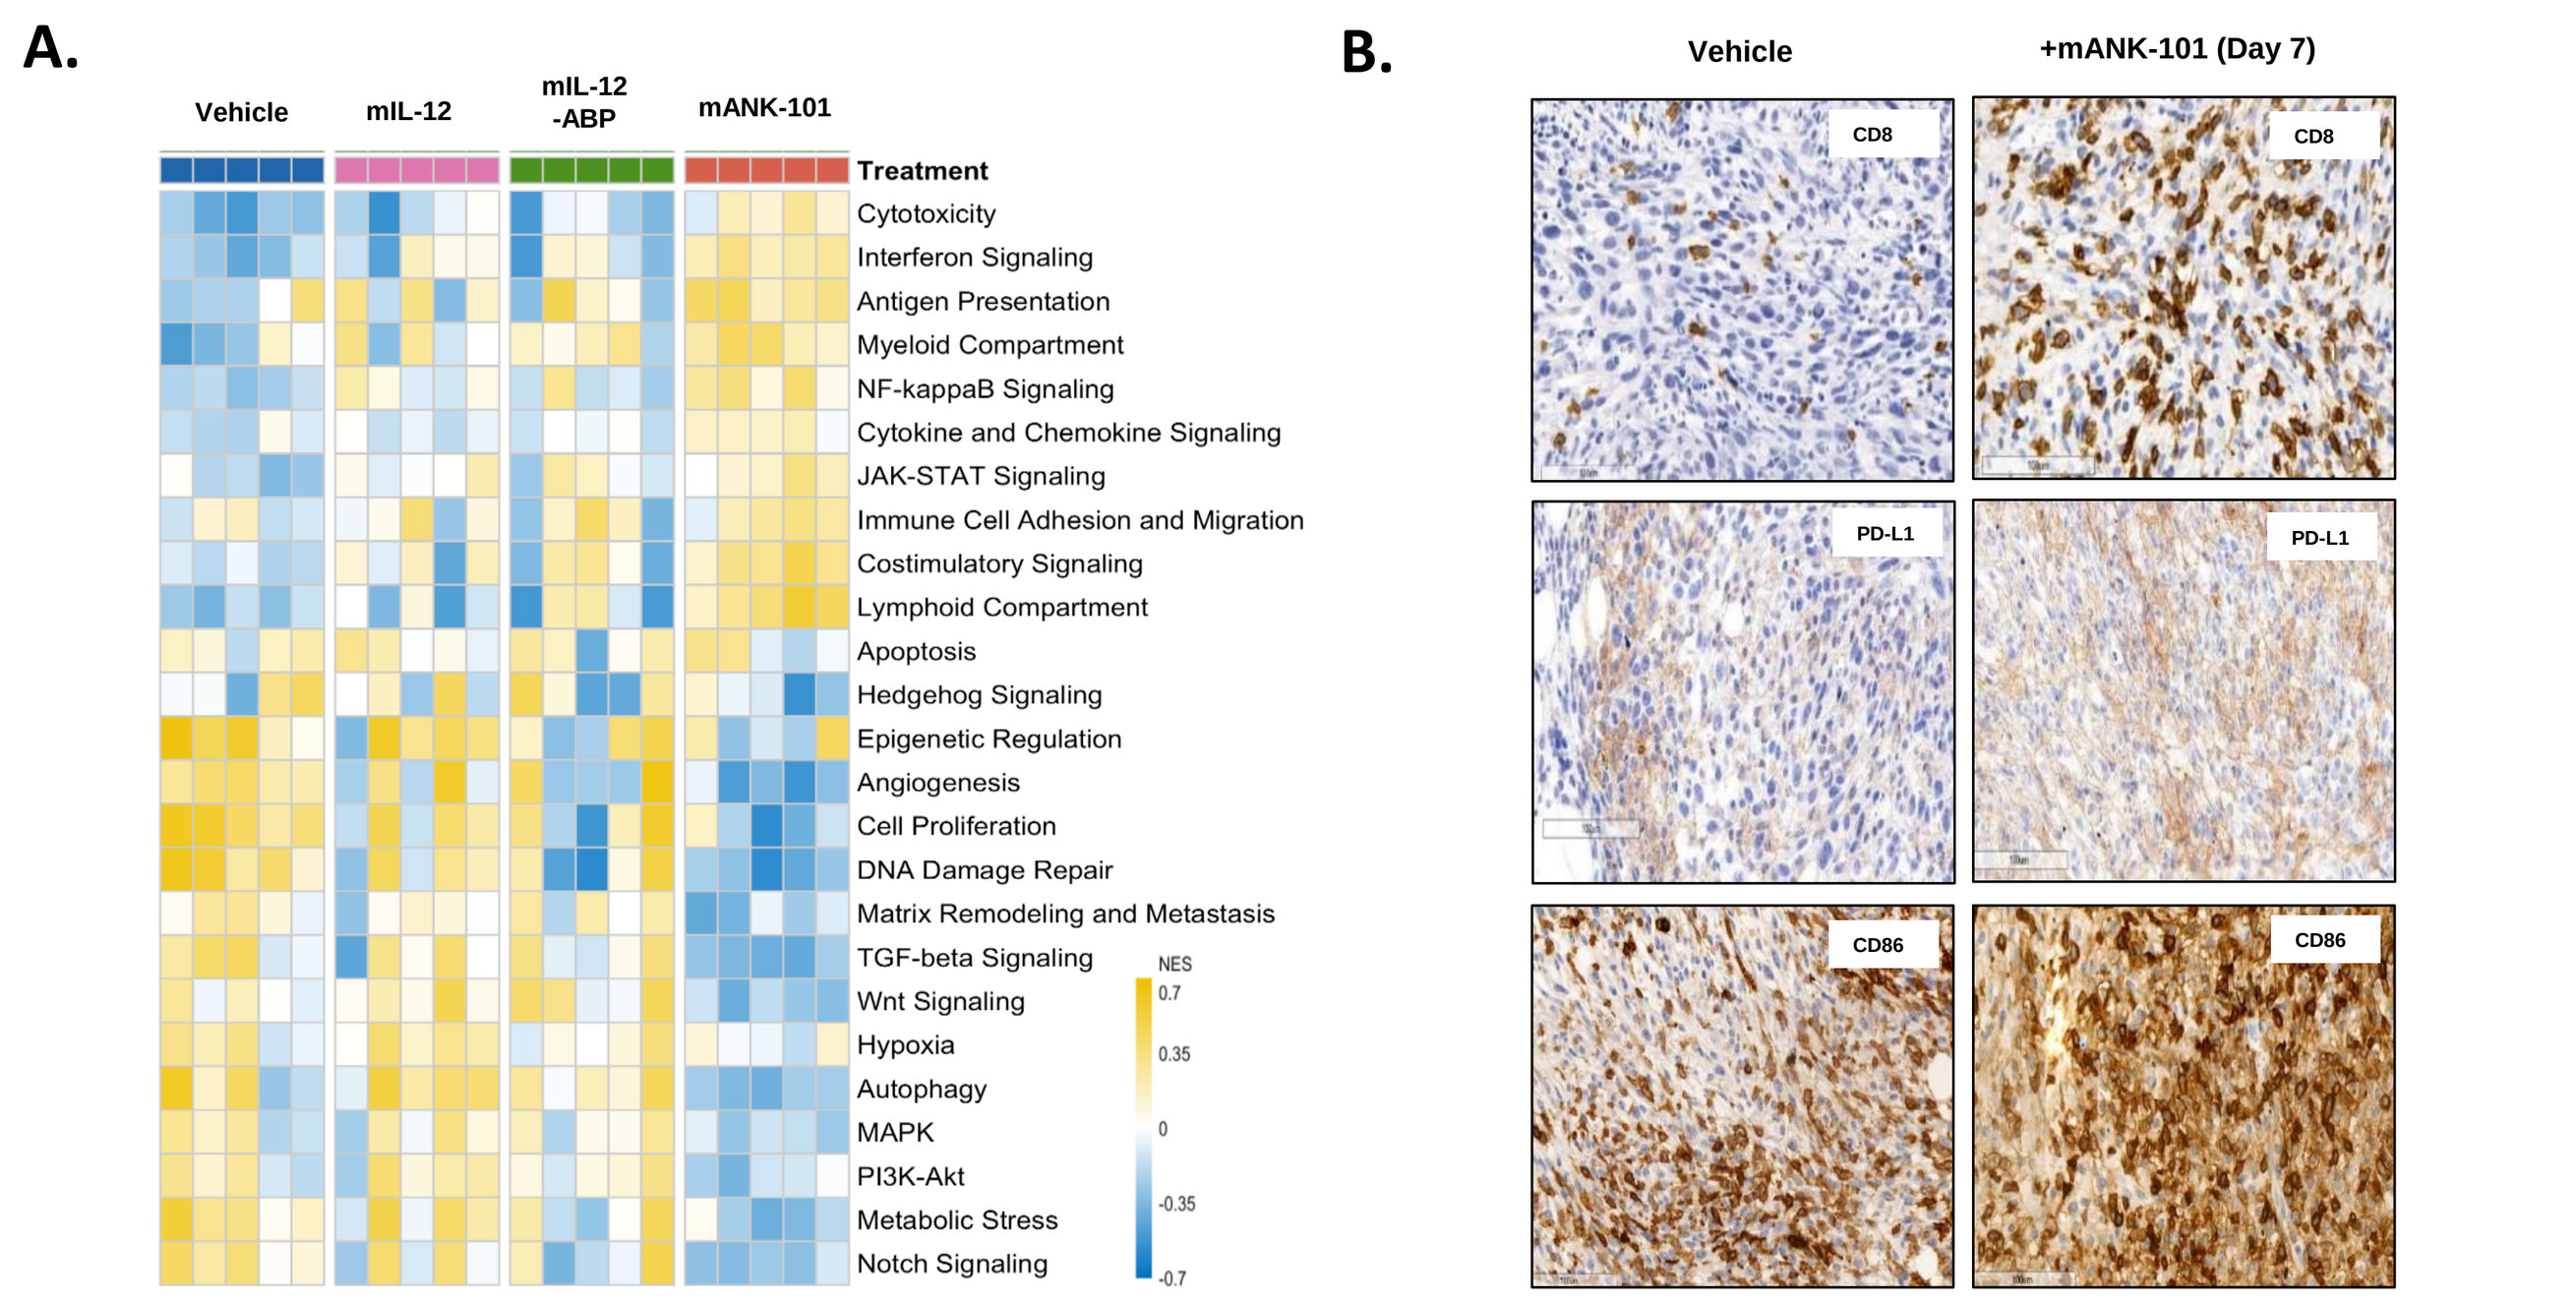

A.
Vehicle
mIL-12
mIL-12 -ABP
mANK-101
Treatment
Cytotoxicity
Interferon Signaling
Antigen Presentation
Myeloid Compartment
NF-kappaB Signaling
Cytokine and Chemokine Signaling
JAK-STAT Signaling
Immune Cell Adhesion and Migration
Costimulatory Signaling
Lymphoid Compartment
Apoptosis
Hedgehog Signaling
Epigenetic Regulation
Angiogenesis
Cell Proliferation
DNA Damage Repair
Matrix Remodeling and Metastasis
TGF-beta Signaling
Wnt Signaling
Hypoxia
Autophagy
MAPK
PI3K-Akt
Metabolic Stress
Notch Signaling
NES
B.
Vehicle
+mANK-101 (Day 7)
CD8
PD-L1
CD86

Figure 7: (A) C57Bl/6 mice bearing MC38 tumors were administered a single IT injection of vehicle or 5 µg of mANK-101. Tumors were harvested 7 days post-treatment and transcriptional analysis was performed by Nanostring (PanCancer IO 360 panel). The data were analyzed using the nSolver analysis software with advanced analysis. Heatmap representing changes in functional annotation pathways with treatment by normalized enrichment score (NES) **(B)** Mice bearing MC38 tumors were treated with a single IT injection of vehicle or 5 µg of mANK-101. On day 7, tumors were excised, fixed, and analyzed by IHC. mANK-101 treated tumors had increased staining for CD8$^+$ T-cells, PD-L1 and CD86 (indicative of myeloid cells transitioning towards an inflammatory M1 phenotype)

Molecular and Cellular Changes with ANK-101 Treatment

Changes in immune modulation consistent with shift to pro-inflammatory Th1 phenotype, NK cell activation and increased antigen presentation at the cellular level

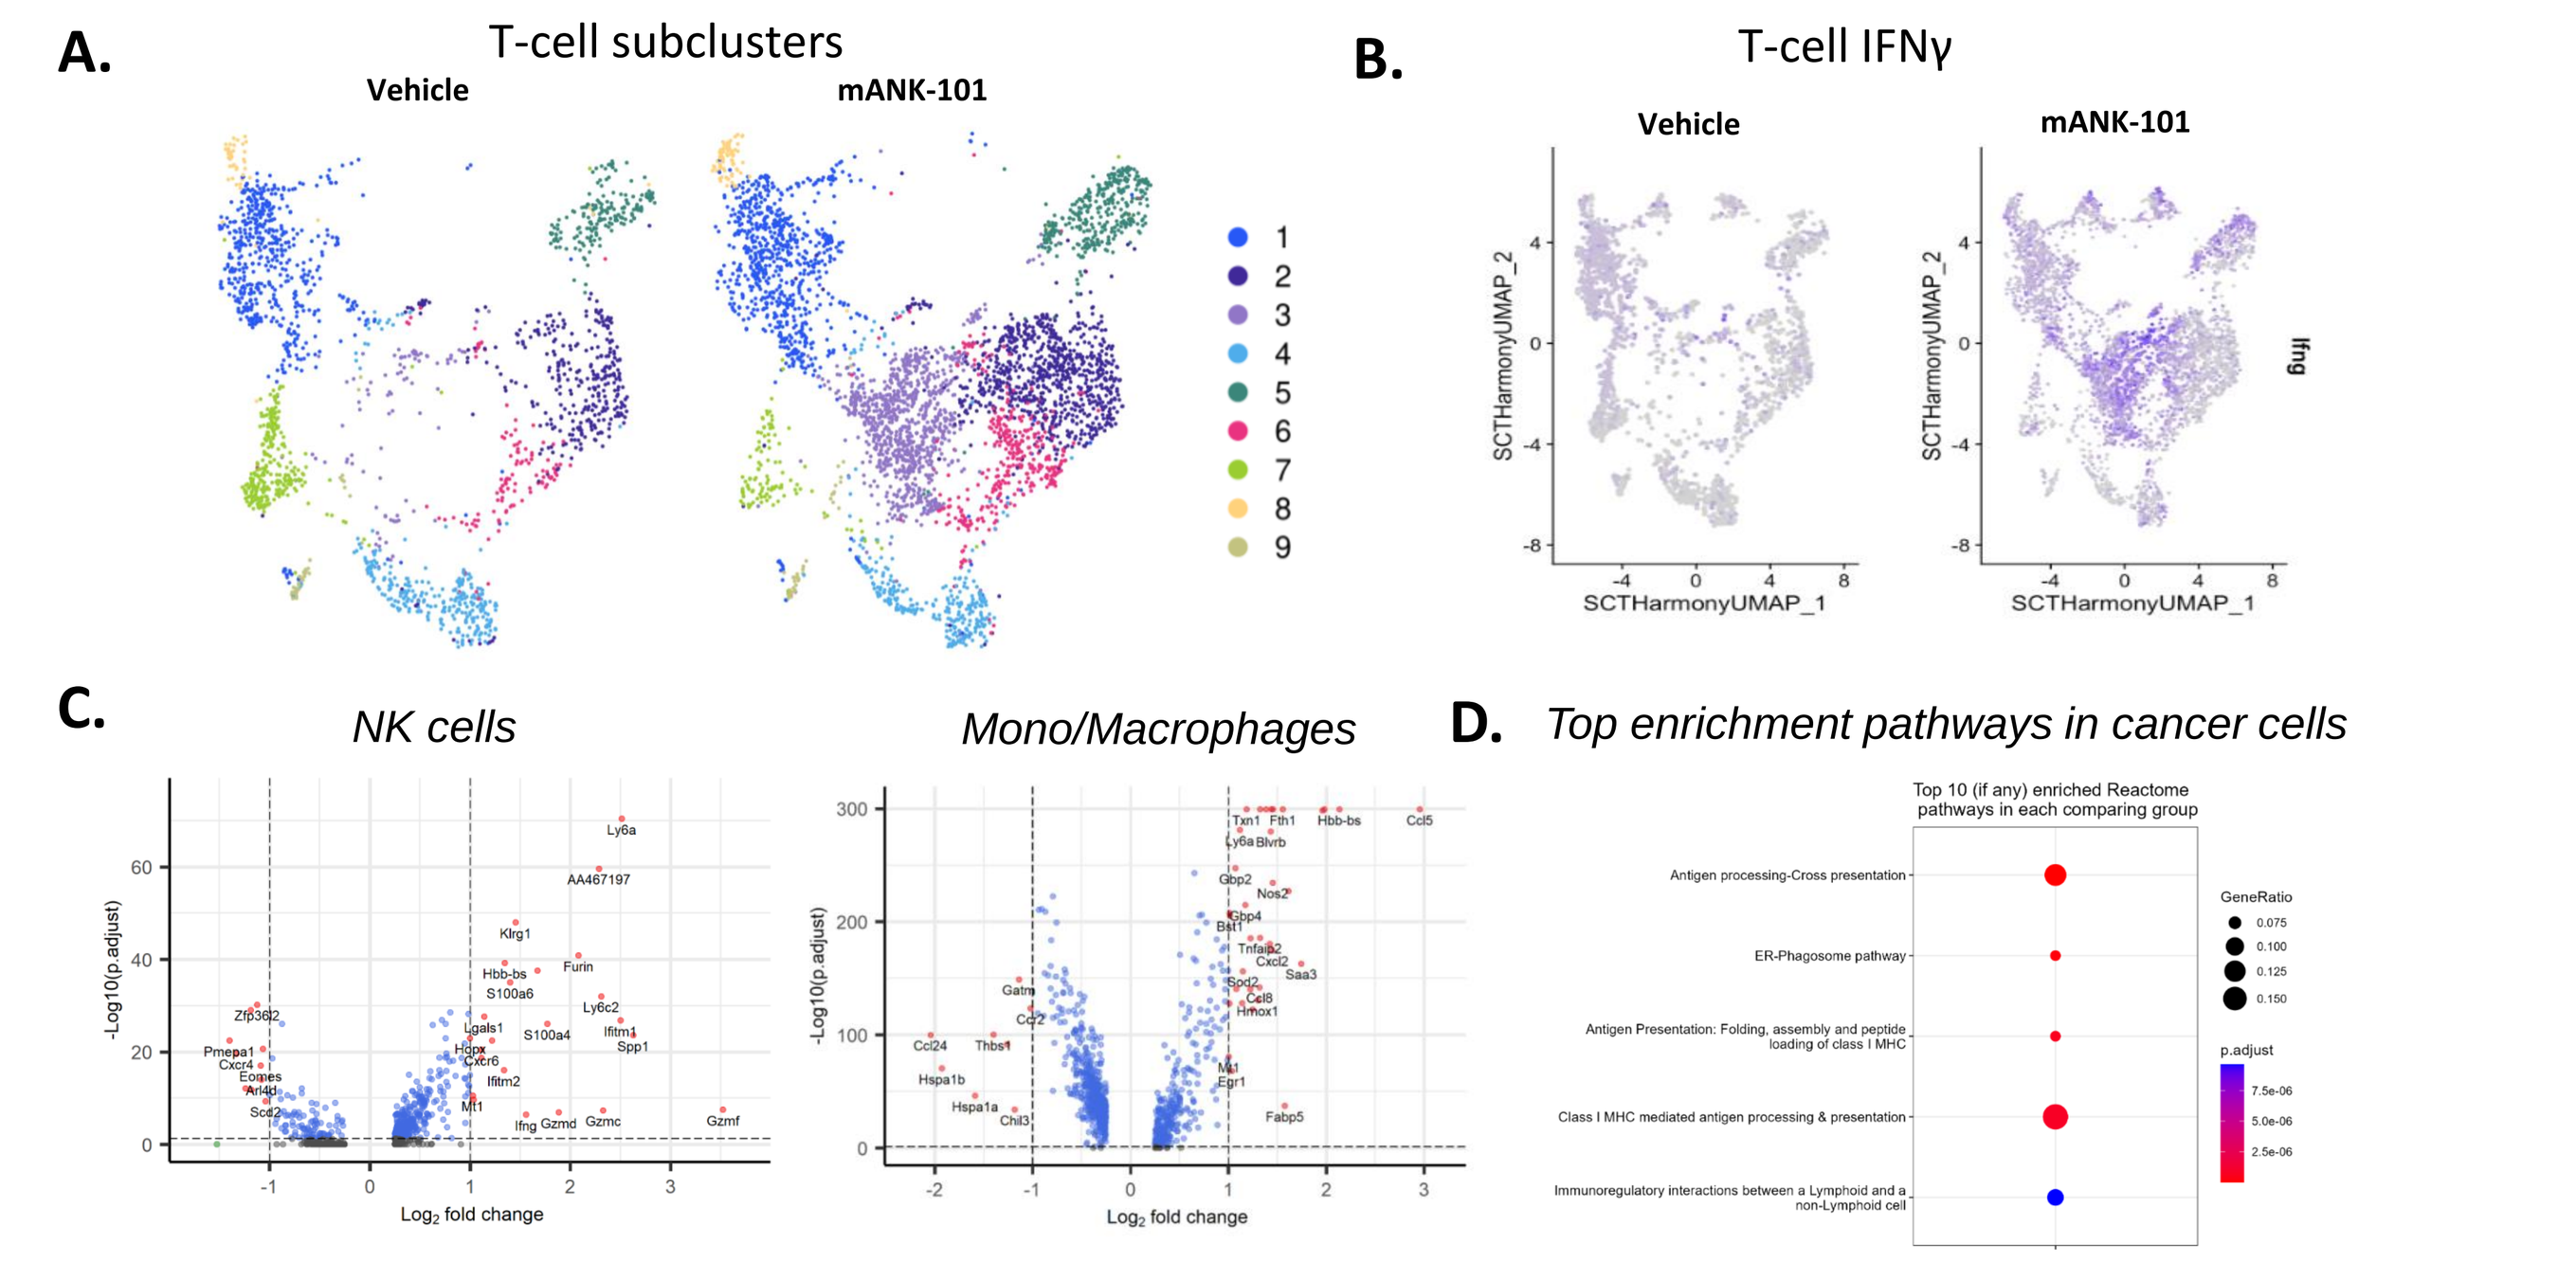

A.
T-cell subclusters
Vehicle
mANK-101
B.
T-cell IFNγ
SCTHarmonyUMAP_1
SCTHarmonyUMAP_2
C.
NK cells
Mono/Macrophages
Log2 fold change
D.
Top enrichment pathways in cancer cells
Antigen processing-Cross presentation
ER-Phagosome pathway
Class I MHC mediated antigen processing & presentation

Figure 8: C57Bl/6 mice bearing MC38 tumors (4 per group) were administered a single IT dose of vehicle or 5 µg of mANK-101, tumors harvested 7 days post-treatment and scRNA-seq was performed according to 10x Genomics protocols. (**A**) UMAP embedding of T lymphocytes colored by cluster identity and by (**B**) expression of IFNγ. mANK-101 treatment induced significant decreases in T-cell cluster 7 (Tregs) and increases in cluster 3 (Th1-like CD4$^+$ cells) (**C**) Volcano plot showing differentially expressed genes with ANK-101 treatment in NK cells (left), mono/macrophage populations (right). (**D**) Top enriched pathways in the cancer cells show increased antigen presentation pathways

ANK-101 is well tolerated in NHPs

ANK-101 is well tolerated in cynomolgus monkeys at all tested dose levels

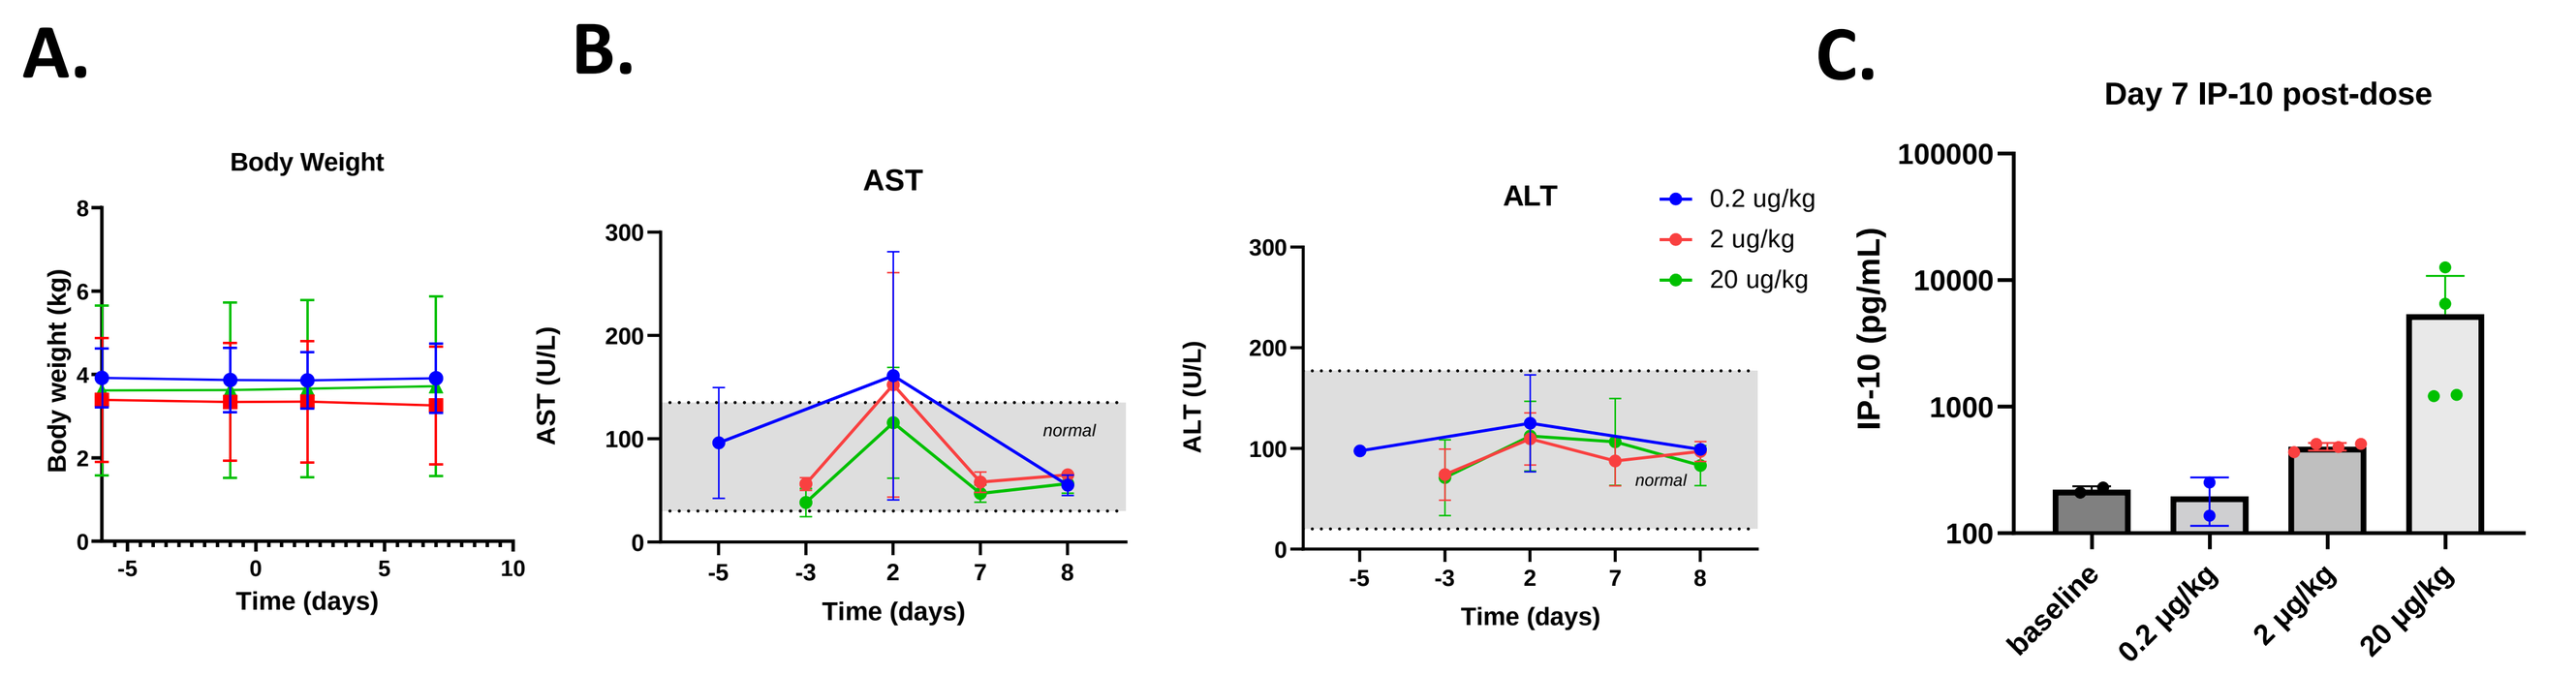

A.
Body Weight
Body weight (kg)
Time (days)
B.
AST
AST (U/L)
ALT
ALT (U/L)
normal
0.2 ug/kg
2 ug/kg
20 ug/kg
C.
Day 7 IP-10 post-dose
IP-10 (pg/mL)
baseline
0.2 µg/kg
2 µg/kg
20 µg/kg

Figure 9: Cynomolgus monkeys (n=2 per dose group) were dosed 0.2, 2, 20 µg/kg SC on Day 1 in single dose group and 2 and 20 µg/kg on day 1 and 8 in the repeat dose groups. There were no clinical observations or treatment dependent body weight changes **(A)** Clinical chemistry analyses indicated only mild, transient changes in liver enzymes (AST and ALT) after the first dose that fully resolved and were not-dose dependent **(B)**. Other liver enzyme markers were unchanged (data not shown). A dose-dependent increase in circulating IP-10 levels was observed on Day 7 **(C)**, with no increases in systemic IL-1β, IL-2, IL-10 and TNFα levels (data not shown)

Conclusions

- Ankyra's proprietary 'Anchored Immunotherapy' platform utilizes the FDA-approved vaccine adjuvant Alhydrogel® (aluminum hydroxide) as a scaffold to locally retain potent cytokine drugs after IT administration to improve their therapeutic window.
- Ankyra's lead program ANK-101 is a stable complex of a modified IL-12 cytokine with Alhydrogel® that retains IL-12 signaling activity
- Biodistribution studies in mice with ^{125}I labeled mANK-101 surrogate demonstrate significantly enhanced tumor retention and reduced systemic exposure compared to free IL-12-ABP
- mANK-101 is efficacious in diverse syngeneic tumor models, including large tumors, after 1 or 2 injections
- mANK-101 treatment induces a profound remodeling of the tumor microenvironment with increased T- and NK-cell infiltration and activation, shifts to pro-inflammatory myeloid cell phenotypes, and increased markers of antigen presentation and co-stimulation as measured by Nanostring, IHC, and scRNA-seq
- Subcutaneous administration of ANK-101 in cynomolgus macaques at expected clinical doses was well tolerated
- IND enabling studies for ANK-101 are on-going with an IND planned in Q2, 2023
- Ankyra is also developing a pipeline of other differentiated immunomodulatory agents

References: *1. Lasek et al. Cancer Immunol Immunother, 2014; 2. Wittrup KD et.al., Expert Opin Drug Del. 2002; 1–8*

Ankyra Therapeutics
83 Newbury Street, 4th Floor
Boston, MA 02116
www.ankyratx.com